N. 20A157 Pathogenic, glycolytic PD-1+ B cells accumulate in the hypoxic RA joint.

Floudas A[1], Neto N[2,3], Marzaioli V[1,4], Murray K[4], Moran B[5], Monaghan M.G[2,3], Low C[4], Mullan RH[6], Rao NL[7] , Krishna V[7], Nagpalr S[7], Veale DJ[4], Fearon U[1,4].

[1] Molecular Rheumatology, Trinity Biomedical Sciences Institute, TCD. [2]Department of Mechanical and Manufacturing Engineering, TCD. [3]Trinity Centre for Biomedical Engineering, TCD. [4]EULAR Centre of Excellence, Centre for Arthritis and Rheumatic Diseases, St Vincent's University Hospital, UCD. [5]School of Biochemistry and Immunology, Trinity Biomedical Sciences Institute, TCD. [6] Dept. of Rheumatology, Tallaght University Hospital. [7] Janssen Research & Development, Immunology.

Trinity College Dublin
Coláiste na Tríonóide, Baile Átha Cliath
The University of Dublin

eular Centres of Excellence

card

Introduction.

Rheumatoid arthritis (RA) often has a progressive and debilitating course, with significant impact on the patient's quality of life. Despite the long-known association with autoantibodies, knowledge of the role of B-cells and their potential direct contribution to RA pathogenesis remains elusive.

Objectives.

Characterization of B cell subpopulation in the periphery and synovial tissue.

Identification of chemokines and their receptors responsible for the accumulation of invading B cells in the inflamed joint.

Characterization of B cell function and metabolism under the hypoxic conditions of the RA joint.

Highlights

A novel pathogenic glycolytic B-cell population at the site of inflammation offering new opportunities for targeted therapeutic intervention in the treatment of RA.

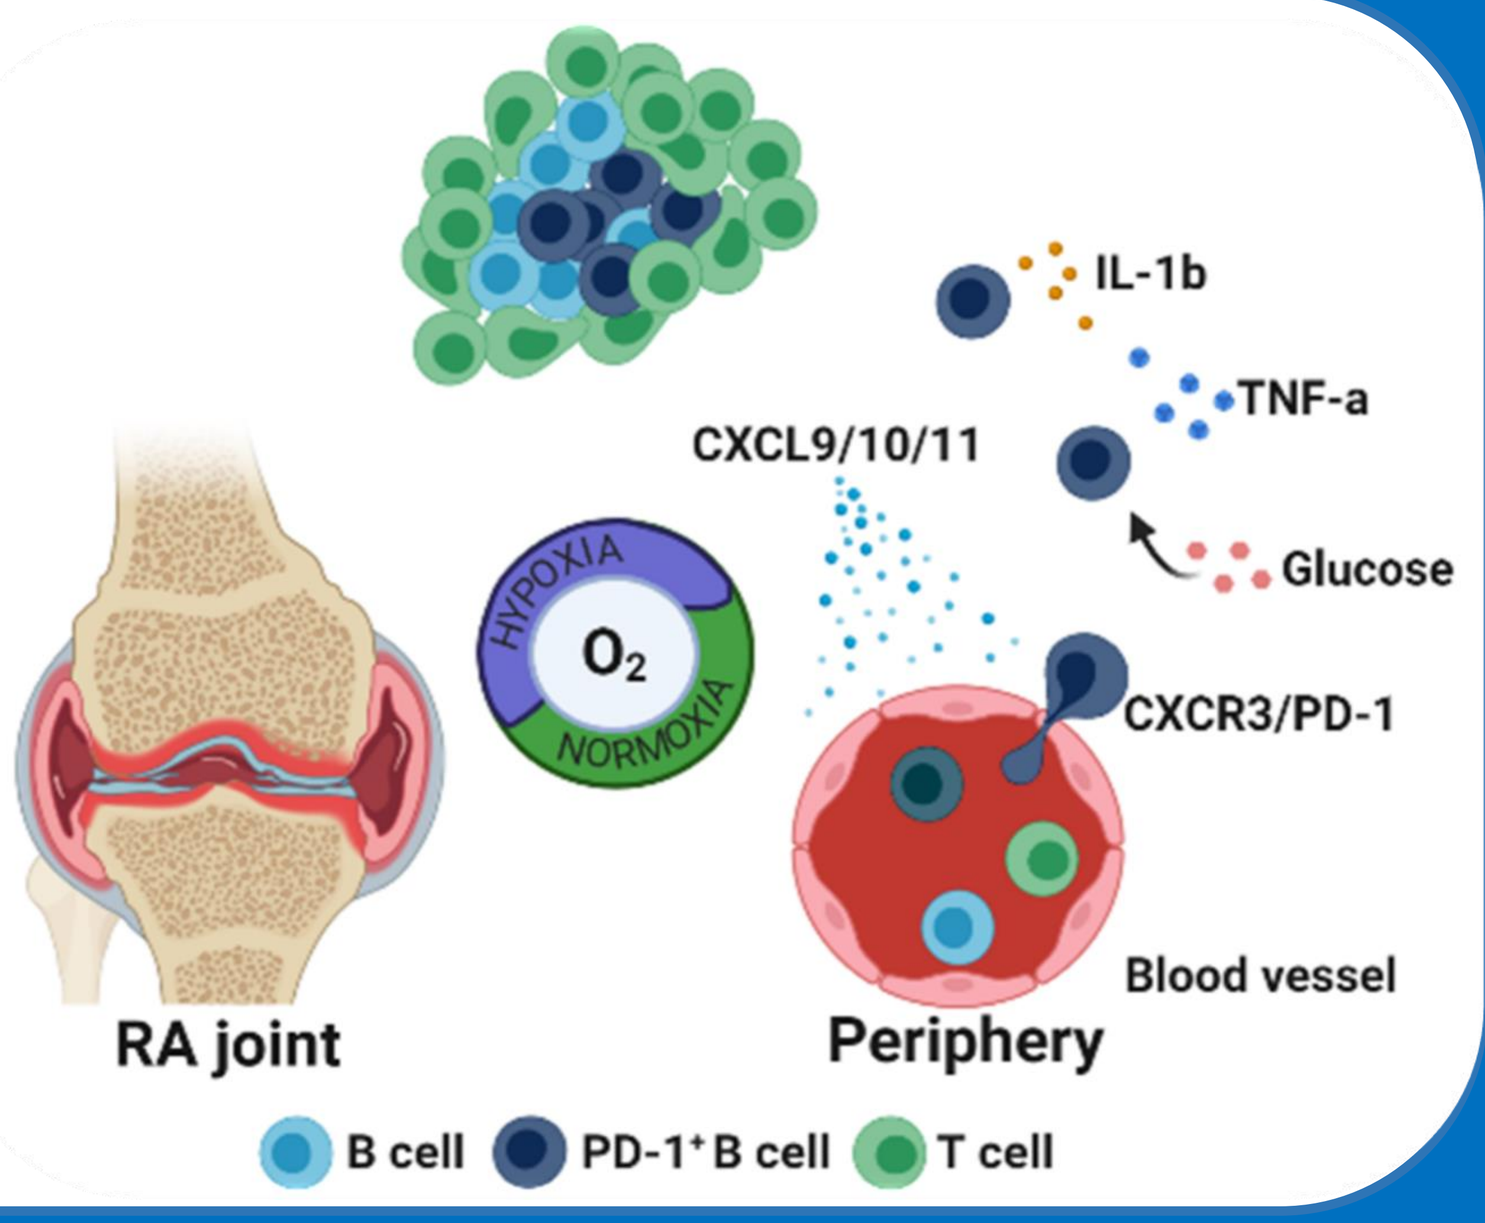
IL-1b
TNF-a
CXCL9/10/11
Glucose
HYPOXIA
O_2
NORMOXIA
CXCR3/PD-1
Blood vessel
RA joint
Periphery
B cell
PD-1+ B cell
T cell

CXCR3 is an important mediator for the synovial accumulation of memory B cells in RA

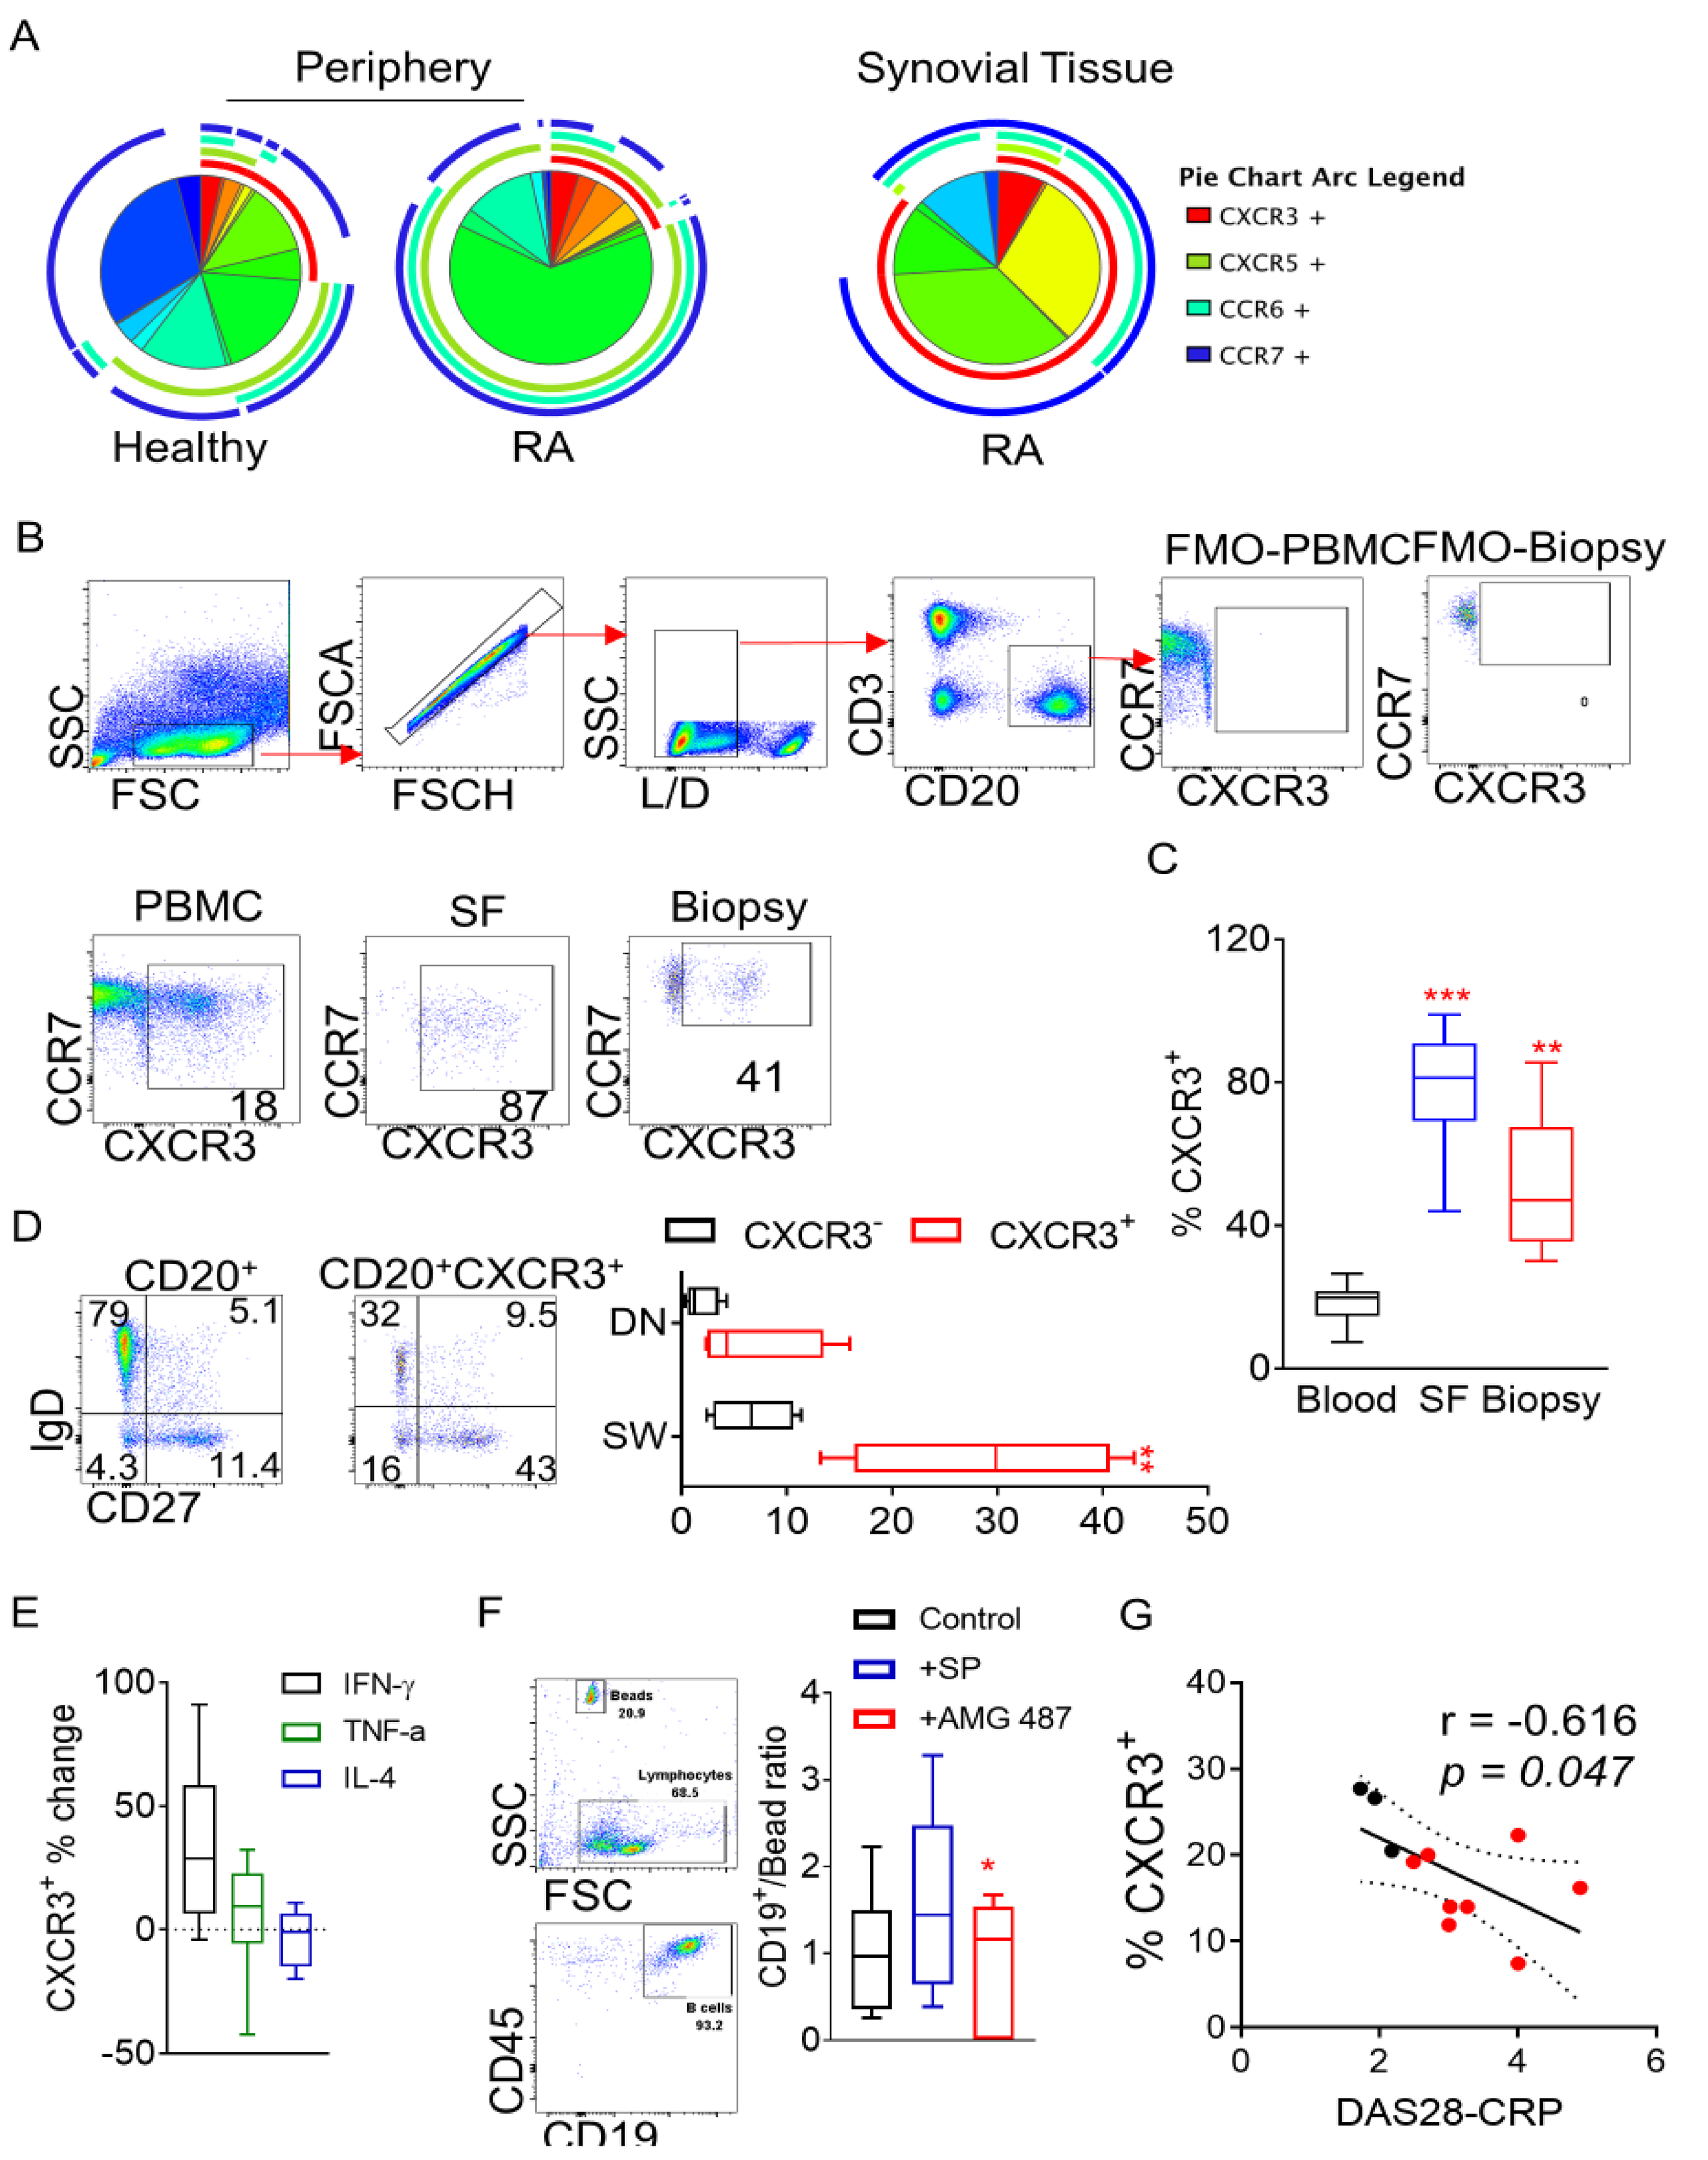
A
Periphery
Synovial Tissue
Pie Chart Arc Legend
CXCR3 +
CXCR5 +
CCR6 +
CCR7 +
Healthy
RA
RA
B
FMO-PBMC
FMO-Biopsy
SSC
FSC
FSCA
FSCH
L/D
CD3
CD20
CCR7
CXCR3
PBMC
SF
Biopsy
18
87
41
C
% CXCR3+
Blood
SF
Biopsy
D
CD20+
CD20+CXCR3+
IgD
CD27
CXCR3-
CXCR3+
DN
SW
E
IFN-γ
TNF-a
IL-4
CXCR3+ % change
F
Control
+SP
+AMG 487
Beads
Lymphocytes
B cells
SSC
FSC
CD45
CD19
CD19+/Bead ratio
G
r = -0.616
p = 0.047
% CXCR3+
DAS28-CRP

A-C) RA patient synovial fluid and synovial tissue B cells maintain high expression of the chemokine receptor CXCR3. (D) The majority of peripheral blood CXCR3 B cells belong to the switched (IgD-CD27+) and double negative (IgD-CD27-) memory compartments, reflecting the B cell subpopulation distribution seen in the synovial tissue of RA patients. (E) Proinflammatory cytokines, associated with pathology in RA, influence B cell CXCR3 expression. (F) RA patient B cell invasion in response to patient biopsy conditioned media, can be mitigated by the AMG487 inhibitor of CXCR3. (G). Peripheral blood CXCR3 B cell frequency inversely correlates with disease activity. These data highlight a key role for CXCR3 in the accumulation of memory B cells at the site of inflammation in RA.

CXCR3^high B cells constitutively express PD-1 and accumulate in the inflamed joint

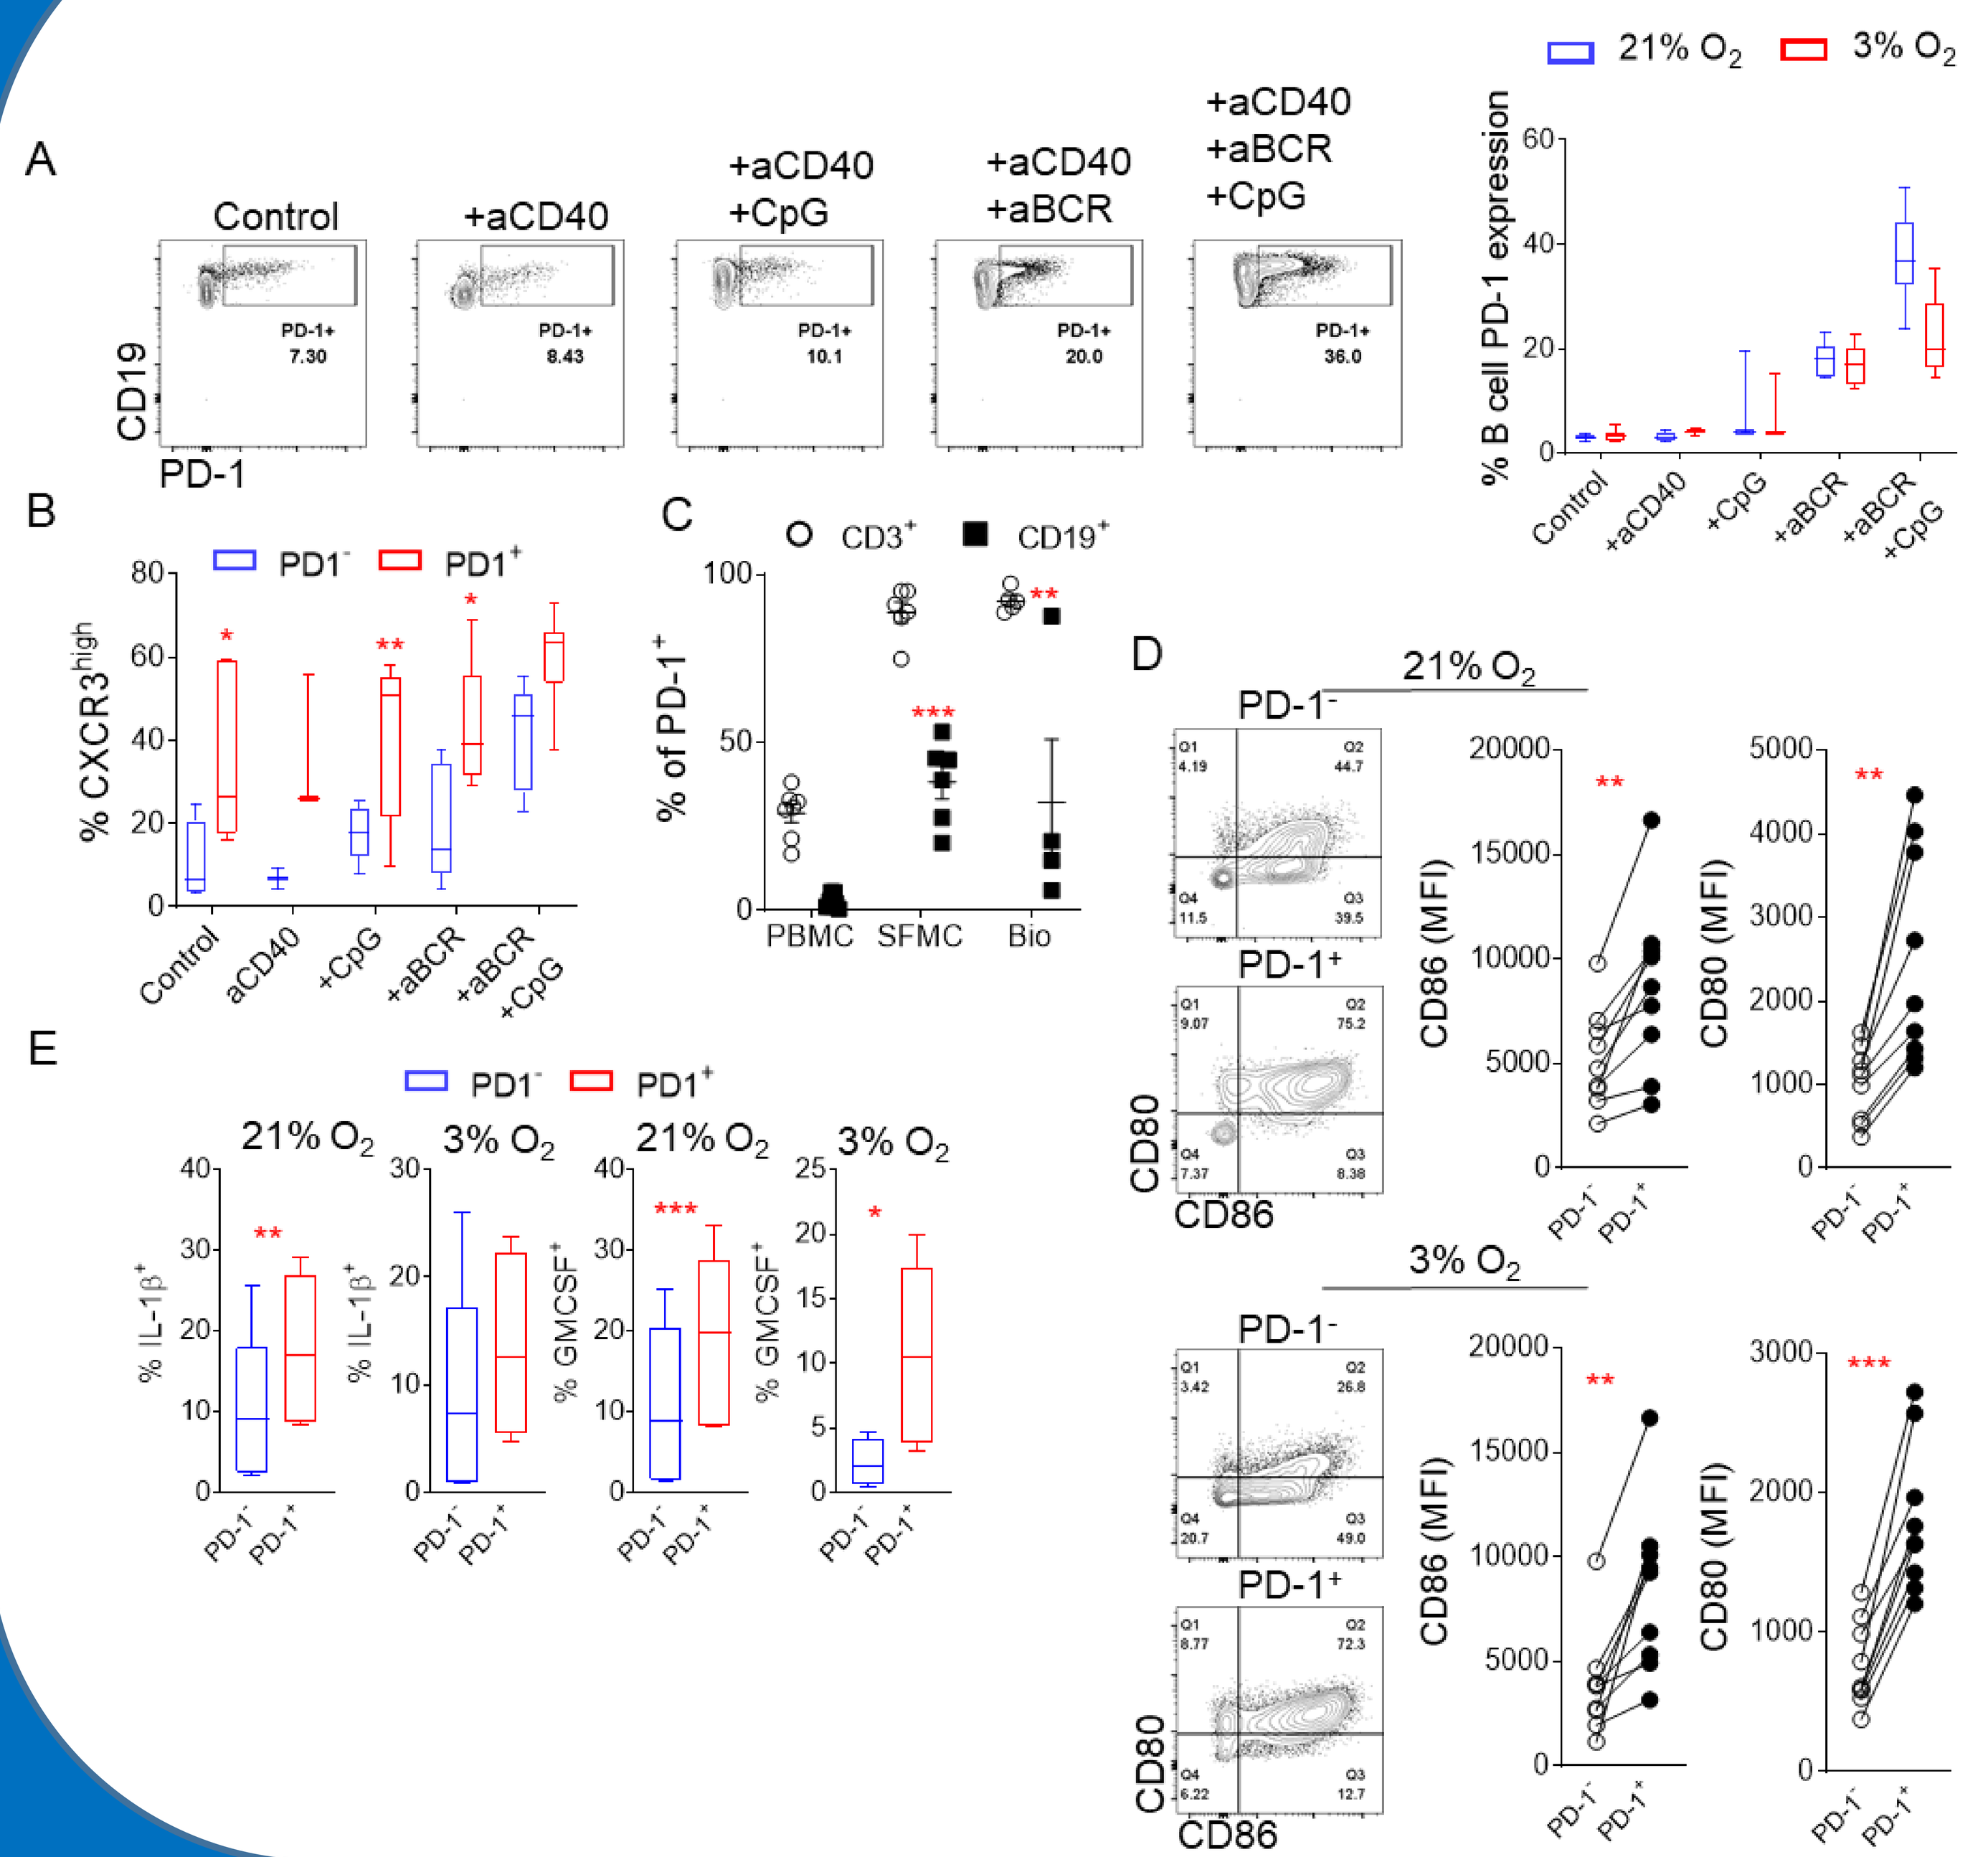
21% O_2
3% O_2
A
Control
+aCD40
+aCD40 +CpG
+aCD40 +aBCR
+aCD40 +aBCR +CpG
CD19
PD-1
% B cell PD-1 expression
B
PD1-
PD1+
% CXCR3high
C
CD3+
CD19+
% of PD-1+
PBMC
SFMC
Bio
D
PD-1-
PD-1+
CD80
CD86
CD86 (MFI)
CD80 (MFI)
E
% IL-1β+
% GMCSF+

(A-B) RA patient B cells were isolated and stimulated in vitro under physiologically relevant stimulation conditions. Importantly B cells were cultured under atmospheric oxygen conditions (normoxia – 21% O_2) or hypoxic conditions (hypoxia – 3% O_2) that simulate the environment of the inflamed joint. CXCR3^high B cells under normoxic and hypoxic conditions, constitutively expressed PD-1 following B cell receptor (BCR) stimulation. (C) PD-1 expressing B cells accumulate in the synovial fluid (~40%) and synovial tissue (~20%) compared to the peripheral blood (~1%). (D, E). PD-1 RA patient B cells maintain higher T cell costimulatory capacity due to increased expression of CD86 and increased proinflammatory cytokine production under normoxic and hypoxic conditions when compared to their PD-1- counterparts.

PD-1+ B cells have higher mTOR pathway activation and depend on glucose uptake

(A,B) RA patient derived PD-1+ B cells stimulated in vitro, show increased activation of the mTOR pathway and subsequently maintain higher expression of the glucose transporter GLUT-1 compared to their PD-1- counterparts. (C) Increase GLUT-1 expression by PD-1+ B cells leads to increased glucose uptake (as demonstrated by uptake of the fluorescent glucose analog 2-NBDG). (D) PD-1+ B cell dependence on glucose is further highlighted by the loss of PD-1 expression following glucose deprivation.

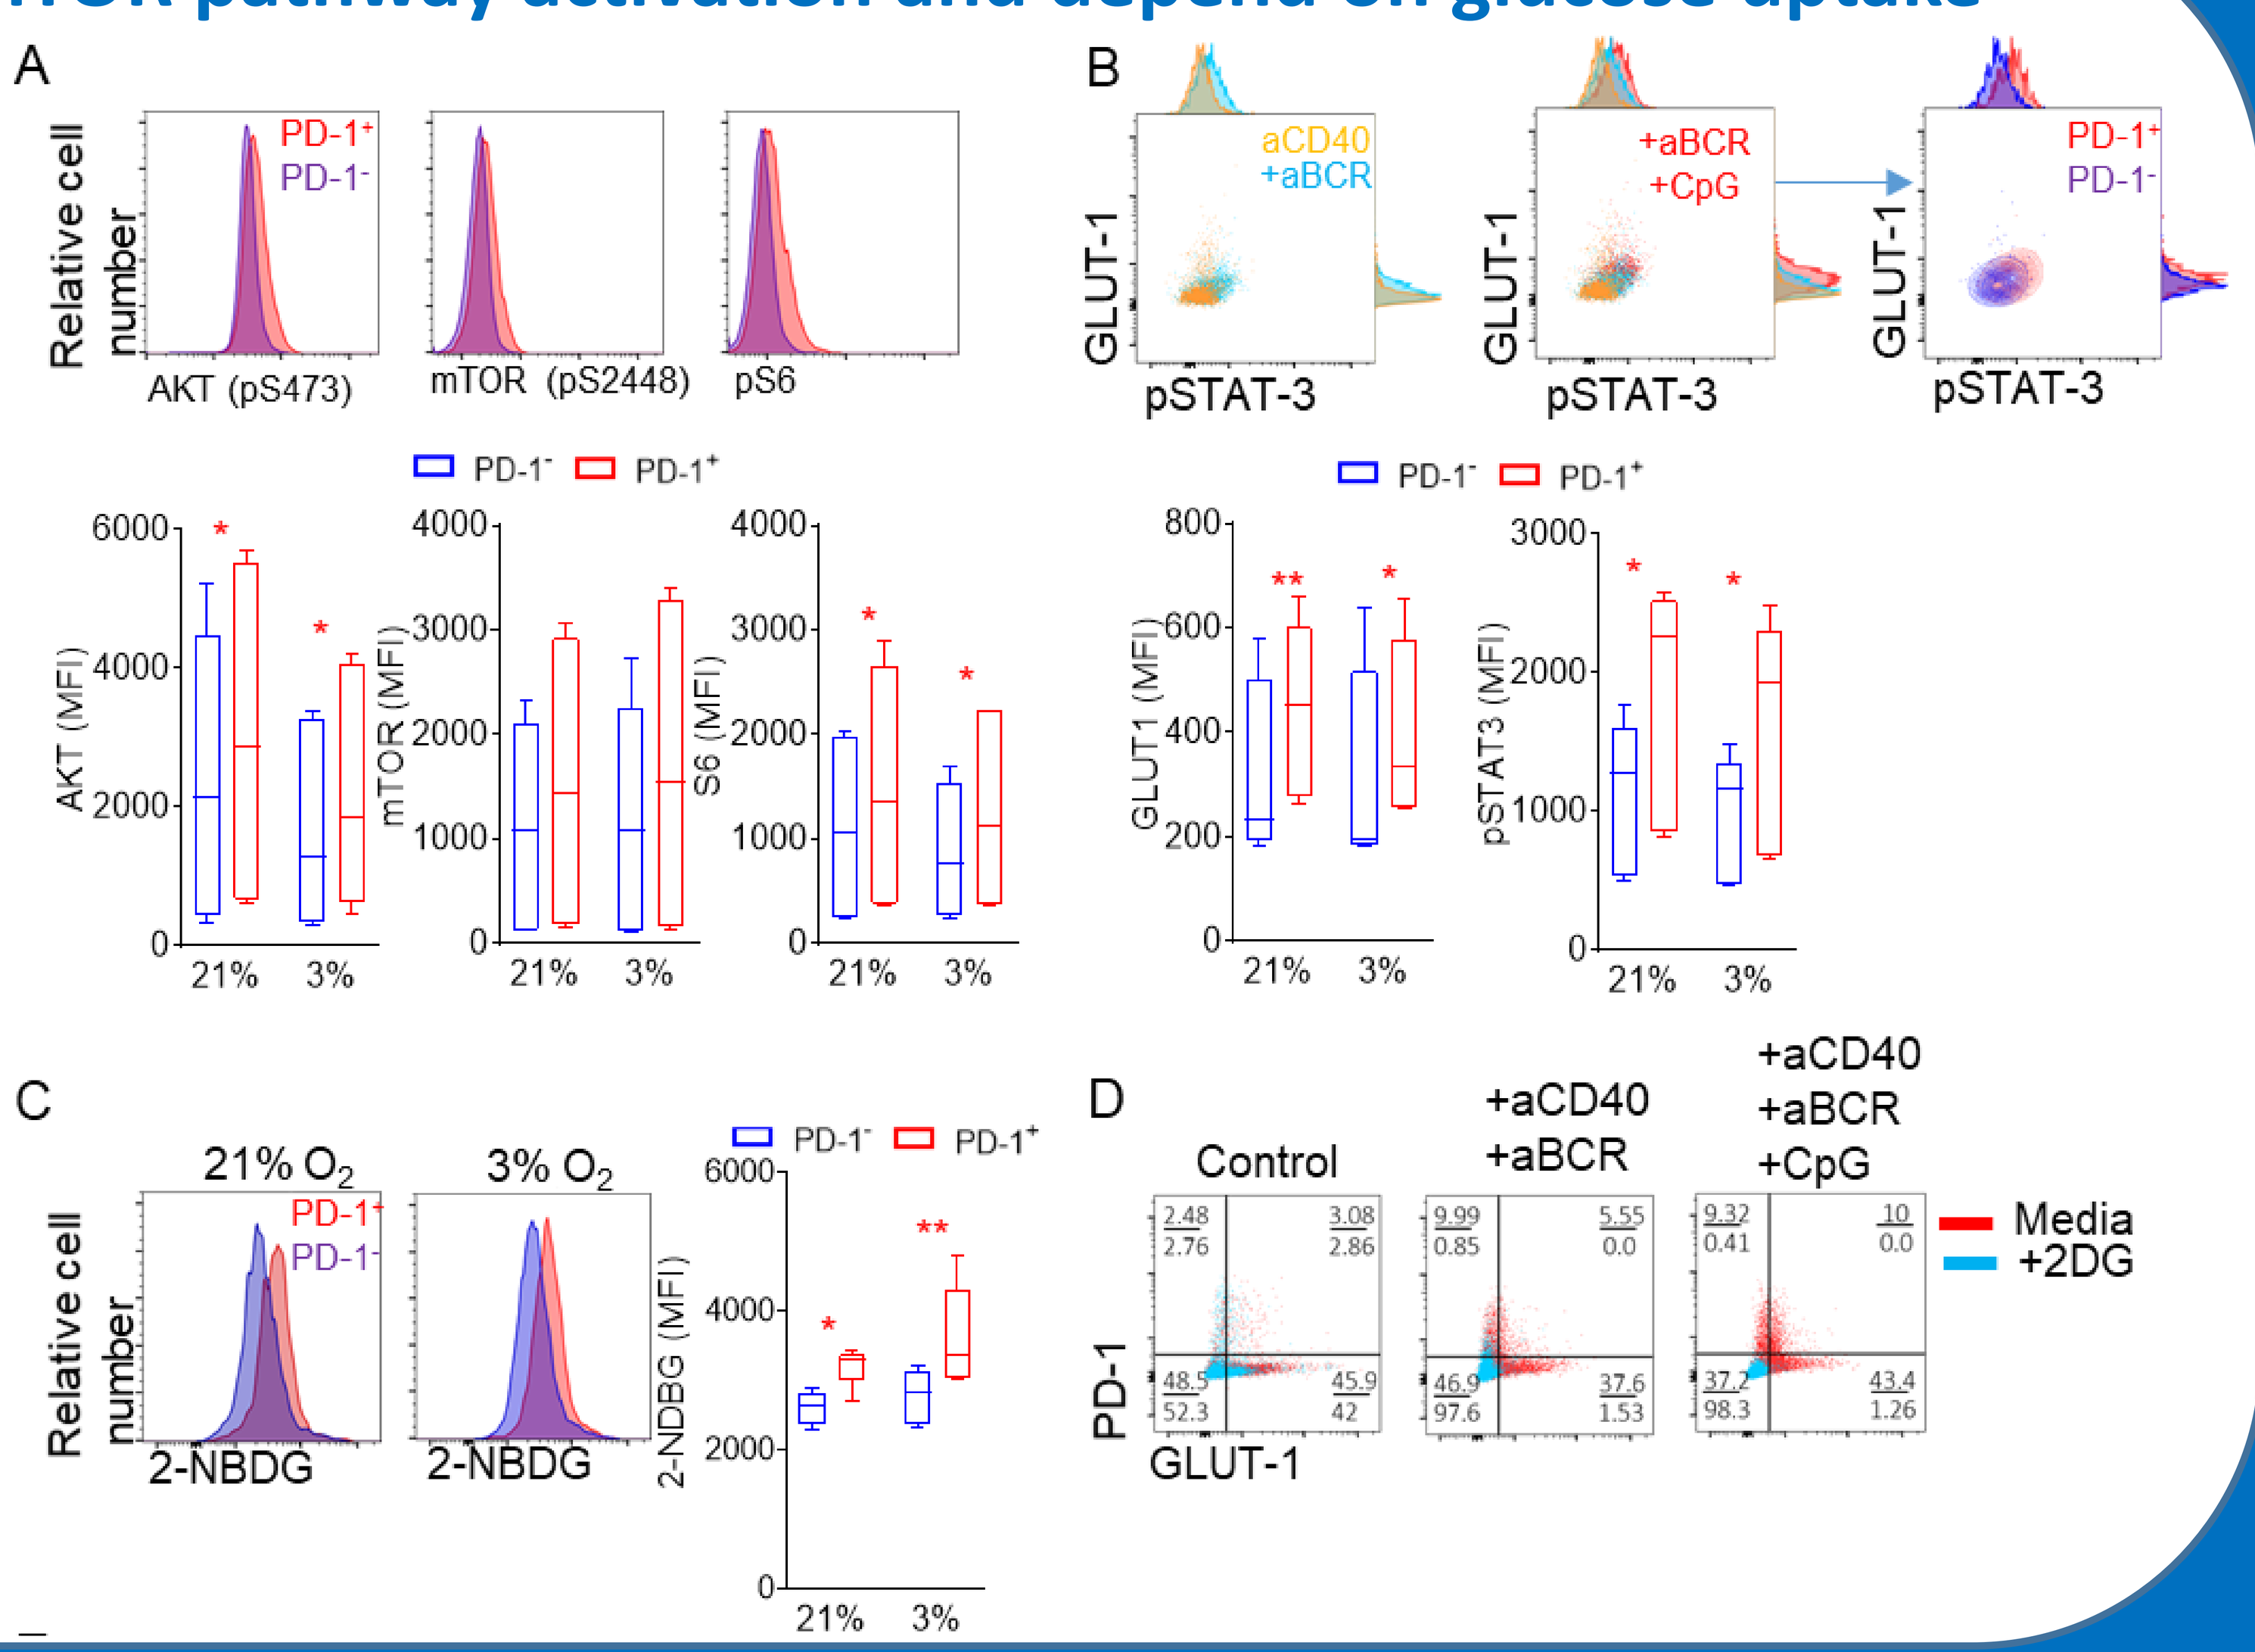
A
Relative cell number
PD-1+
PD-1-
AKT (pS473)
mTOR (pS2448)
pS6
AKT (MFI)
mTOR (MFI)
S6 (MFI)
B
aCD40 +aBCR
+aBCR +CpG
GLUT-1
pSTAT-3
GLUT1 (MFI)
pSTAT3 (MFI)
C
21% O_2
3% O_2
2-NBDG
2-NBDG (MFI)
D
Control
+aCD40 +aBCR
+aCD40 +aBCR +CpG
Media
+2DG
PD-1
GLUT-1

RA patient PD-1+ B cells are glycolytic and localise at synovial ectopic lymphoid organs

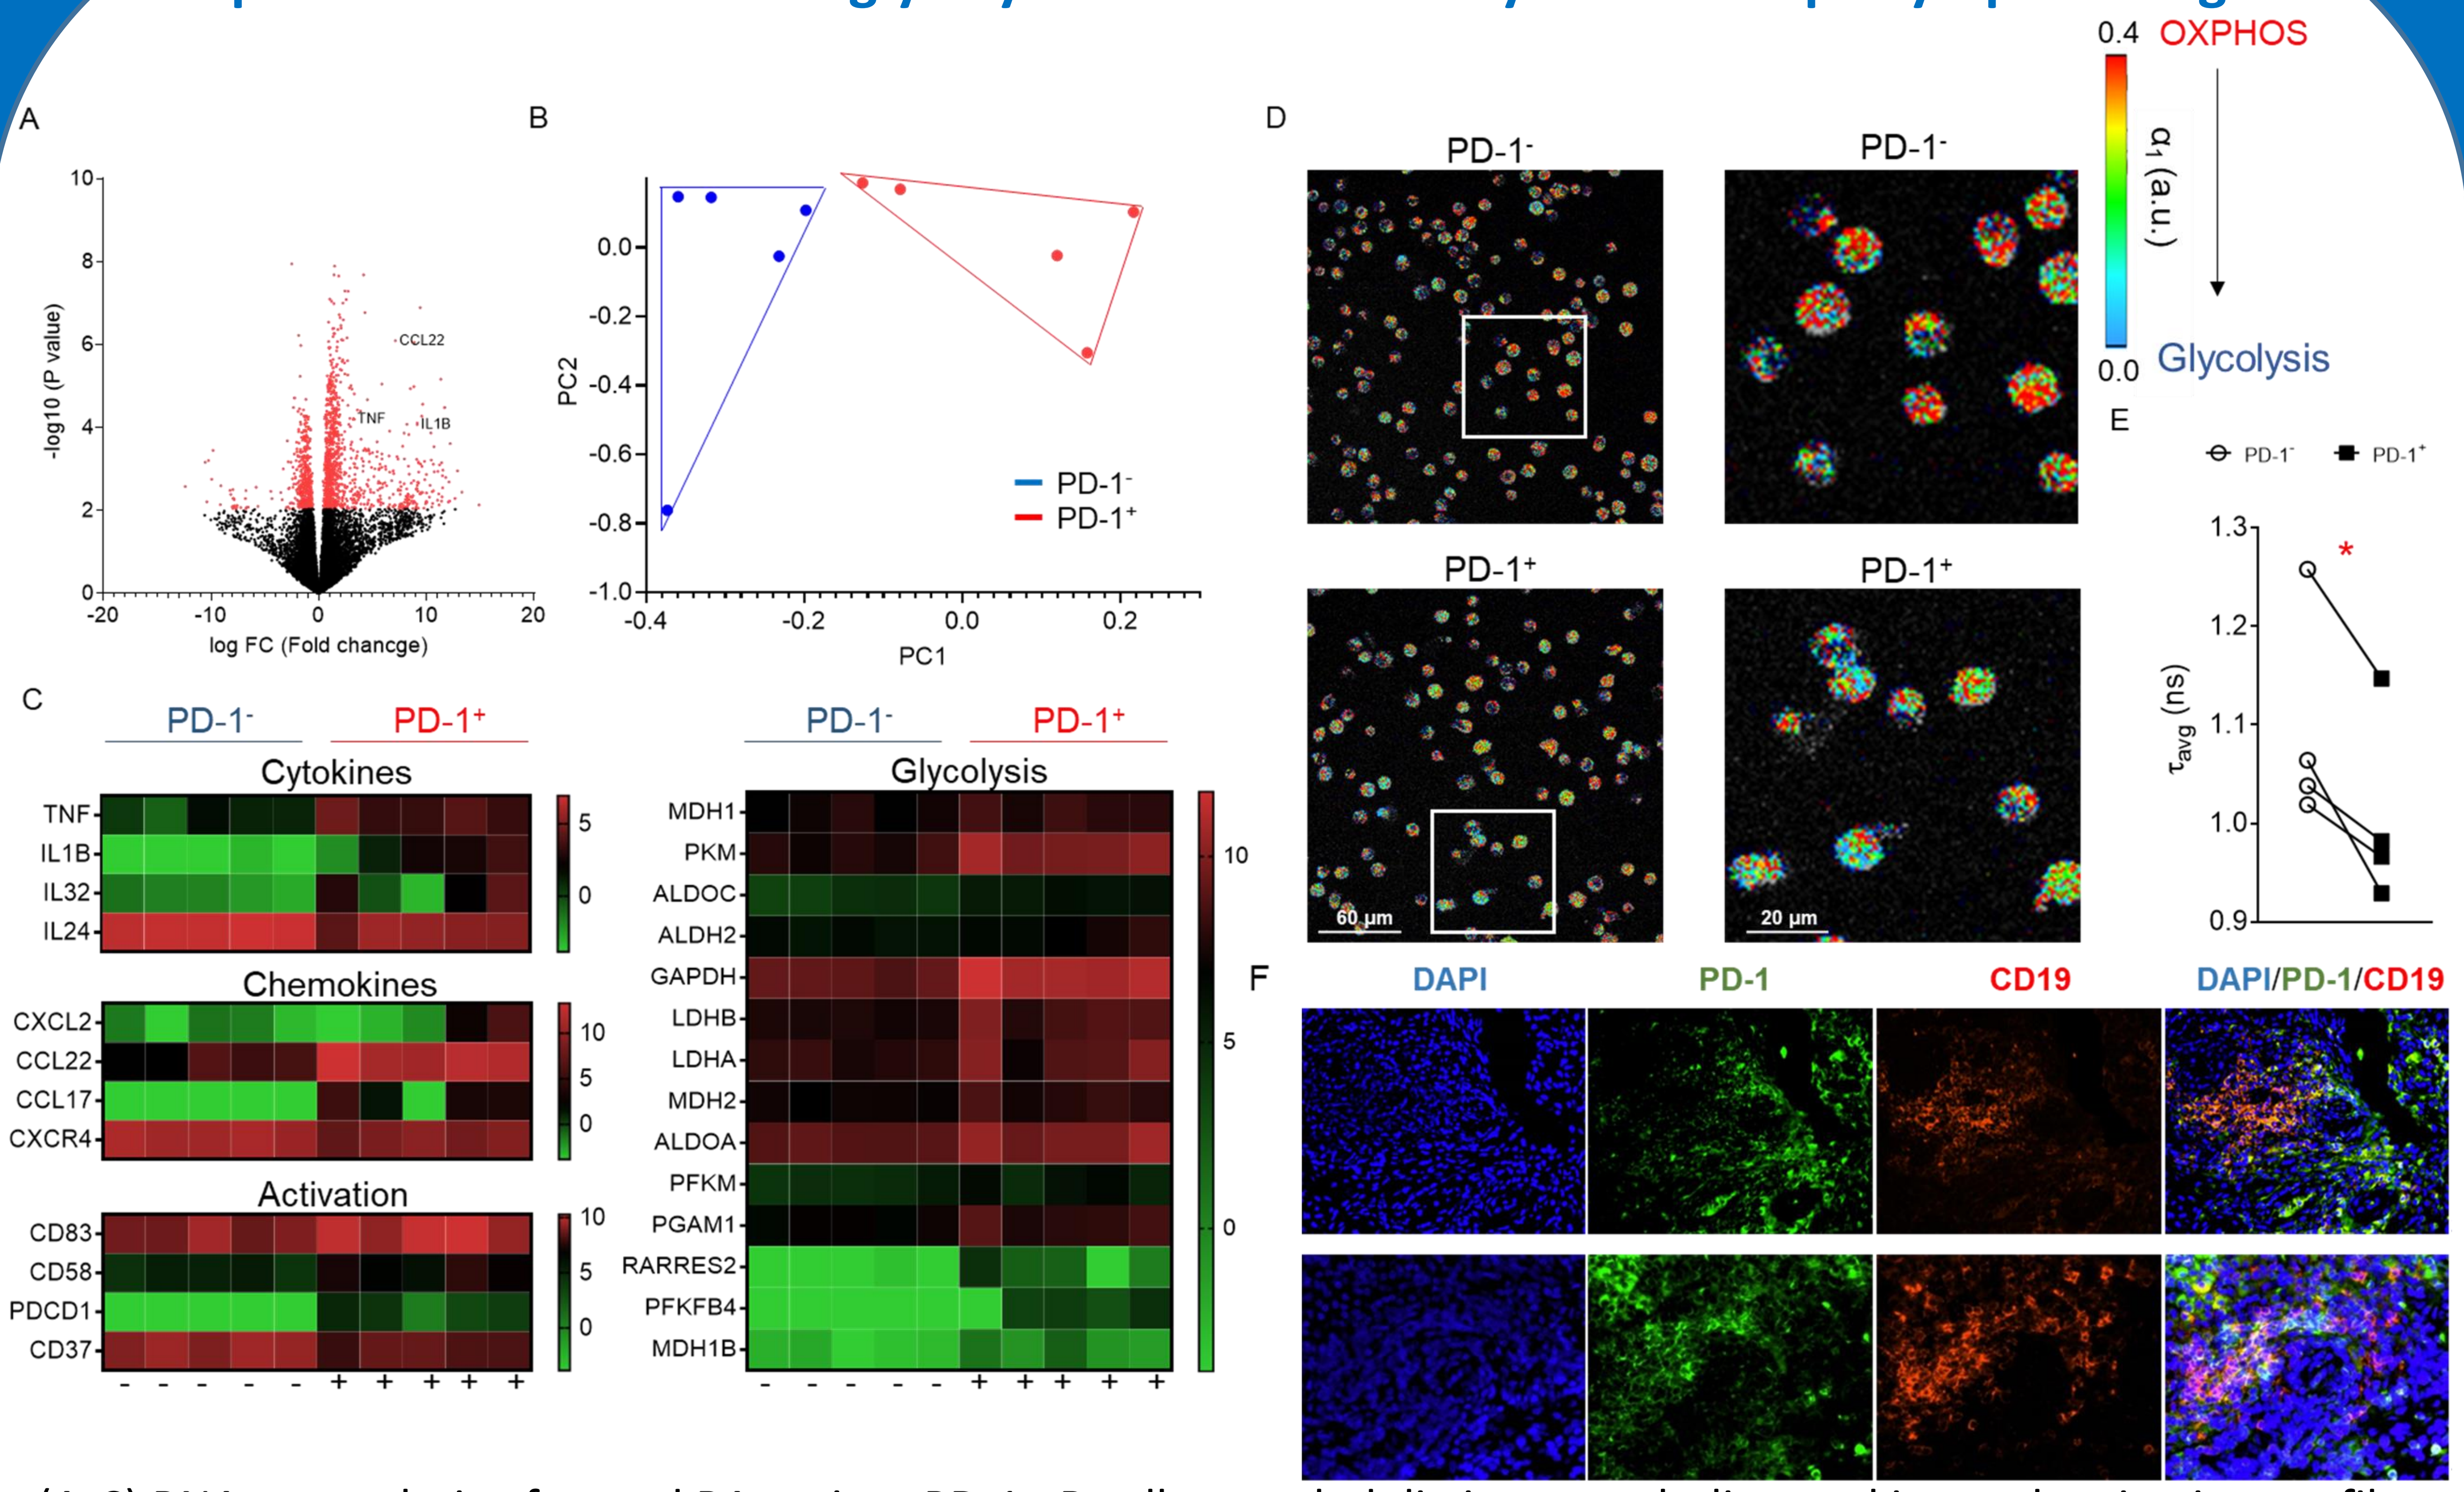
A
-log10 (P value)
log FC (Fold chancge)
CCL22
TNF
IL1B
B
PC2
PC1
PD-1-
PD-1+
C
PD-1-
PD-1+
Cytokines
TNF
IL1B
IL32
IL24
Chemokines
CXCL2
CCL22
CCL17
CXCR4
Activation
CD83
CD58
PDCD1
CD37
Glycolysis
MDH1
PKM
ALDOC
ALDH2
GAPDH
LDHB
LDHA
MDH2
ALDOA
PFKM
PGAM1
RARRES2
PFKFB4
MDH1B
D
PD-1-
PD-1+
60 μm
20 μm
0.4 OXPHOS
α_1 (a.u.)
0.0 Glycolysis
E
τ_{avg} (ns)
F
DAPI
PD-1
CD19
DAPI/PD-1/CD19

(A-C) RNAseq analysis of sorted RA patient PD-1+ B cells revealed distinct metabolic, cytokine and activation profiles. (D) Novel, non-invasive fluorescent imaging microscopy (FLIM) technique highlighted the glycolytic profile of PD-1+ B cells. FLIM allows the visualisation of NADH fluorescence. Importantly, the fluorescent lifetime of NADH is dependent on whether NADH is free or bound. A reduction in τ_{avg} is reflective of an increase in free versus bound NADH which correlates with increased glycolysis over oxidative phosphorylation. (E) These pathogenic, glycolytic PD-1 B cells, preferentially localise at synovial tissue secondary lymphoid organ like structures.

Conclusions.

The accumulation of pro-inflammatory PD-1 B cells in the synovium of RA patients, is CXCR3 mediated and offers an opportunity for early therapeutic intervention. Once in the hypoxic environment of the inflamed RA joint, PD-1 B cells show altered activation, pro-inflammatory cytokine production and metabolism that could prove important for understanding the role of B cells in RA disease pathogenesis.